Efficacy of Sacituzumab Govitecan vs Treatment of Physician's Choice in Previously Treated HR+/HER2– Metastatic Breast Cancer: A Meta-Analysis of TROPiCS-02 and EVER-132-002 Trials

Poster #193P

Oleg Gluz[1], Binghe Xu[2], Rita Nanda[3], Anandaroop Dasgupta[4], Ankita Kaushik[4], Wendy Verret[5], Gianluca Baio[6], Akansha Sharma[7], Barinder Singh[8], Hope S Rugo[9]

[1]Breast Center Niederrhein, Evangelical Hospital Johanniter Bethesda, Mönchengladbach, Germany; [2]Cancer Hospital Chinese Academy of Medical Sciences, Beijing, China; [3]University of Chicago Medicine, Chicago, IL, USA; [4]Health Economics Outcomes Research, Gilead Sciences, Inc., Foster City, CA, USA; [5]Clinical Development, Gilead Sciences, Inc., Foster City, CA, USA; [6]University College London, London, UK; [7]Health Economics Outcomes Research, Pharmacoevidence, SAS Nagar, Mohali, India; [8]Health Economics Outcomes Research, Pharmacoevidence, London, UK; [9]Helen Diller Family Comprehensive Cancer Center, University of California, San Francisco, CA, USA

Copies of this poster obtained through QR, AR, and/or text key codes are for personal use only and may not be reproduced without written permission of the authors.

Conclusions

- This meta-analysis of the global pivotal TROPiCS-02 and EVER-132-002 phase 3 trials showed that SG significantly improved OS and PFS vs TPC in the overall and CDK4/6i-pretreated HR+/HER2– mBC populations
- These results are consistent with trial-level results from TROPiCS-02 and EVER-132-002, reinforcing the efficacy benefits of SG over TPC

Plain Language Summary

- Sacituzumab govitecan is a drug that is approved in several countries to treat hormone receptor–positive/human epidermal growth factor receptor-2–negative (HR+/HER2–) metastatic breast cancer (a type of breast cancer that has spread to other parts of the body), the most common subtype of breast cancer
- In the TROPiCS-02 and EVER-132-002 phase 3 clinical trials, participants with HR+/HER2– metastatic breast cancer received either sacituzumab govitecan or a treatment of physician's choice
- This study is a meta-analysis of the above trials. Meta-analyses typically combine and summarize data from 2 or more trials
- Participants in both trials had similar characteristics except that:
 - All participants in the TROPiCS-02 trial and only about half of the participants in the EVER-132-002 trial had received prior cyclin-dependent kinase 4/6 inhibitors (CDK4/6i, a class of drugs used to treat breast cancer)
 - Most participants in the TROPiCS-02 trial were non-Asian and all participants in the EVER-132-002 trial were Asian
- The results of this meta-analysis showed that participants who took sacituzumab govitecan had more time before their tumor got bigger or spread further or they died from any cause (progression-free survival), and they lived longer (overall survival) compared with participants who took chemotherapy
 - Participants benefited from sacituzumab govitecan whether they had previously been given a CDK4/6i or not

Introduction

- Sacituzumab govitecan (SG) is a Trop-2–directed antibody-drug conjugate that delivers a topoisomerase inhibitor in solid tumors including HR+/HER2– metastatic breast cancer (mBC)[1,2]
- TROPiCS-02 (T-02) is an open-label, randomized, phase 3 trial that demonstrated the efficacy and manageable safety profile of SG vs treatment of physician's choice (TPC) in patients with HR+/HER2–, locally advanced, inoperable, or mBC, after failure of 2 to 4 prior chemotherapy regimens for metastatic disease[2,3]
 - Based on these data, SG is approved in multiple countries worldwide for the treatment of HR+/HER2– mBC following endocrine therapy and at least 2 additional systemic therapies in the metastatic setting[4,5]
- EVER-132-002 (E-002) is another open-label, randomized, phase 3 trial with similar results in this patient population[6]
- T-02 and E-002 had several differences:
 - T-02, the pivotal global trial, enrolled predominantly non-Asian patients, whereas the E-002 trial enrolled only Asian patients
 - T-02, but not E-002, required prior cyclin-dependent kinase 4/6 inhibitor (CDK4/6i) treatment
- The objective of this meta-analysis is to evaluate the efficacy of SG vs TPC in all patients (overall population) and the subset of patients pretreated with CDK4/6i from T-02 and E-002

Methods

- A feasibility assessment of the meta-analytic framework was conducted by evaluating the level of availability of evidence, trial heterogeneity, and approaches to pool effect sizes of SG vs TPC from trials existing in the evidence inventory (Figure 1)
- Meta-analytic models were developed to adjust for cross-trial differences. A one-stage model (results yielded by pooling individual patient level data from T-02 and E-002) and a fixed-effect model (results yielded by pooling trial-level hazard ratios from T-02 and E-002) estimated pooled treatment effects for overall survival (OS) and progression-free survival (PFS) in the overall and CDK4/6i-pretreated populations
 - The one-stage model is the model of choice for enhancing statistical power and precision when estimating treatment effects
 - The fixed-effect model is the model of choice after taking into consideration cross-trial differences (in this case homogeneity between trials being statistically established) when estimating treatment effect
 - The fixed-effect model provides more precise estimates of the treatment effect, especially when the studies are similar in design, population, and variation in treatment outcomes. The fixed-effect model was selected as the base-case model due to nonsignificant Chi² statistic, moderate heterogeneity (< 75% I^2 statistic), and expert guidance
 - The difference in patient geography between the 2 trials cannot be adjusted, but prior studies have suggested that this difference has no significant impact on survival outcomes in breast cancer
- A regression analysis was conducted with respect to the various clinical characteristics, and the significant heterogenous covariates identified during the assessment were adjusted in the meta-analysis (Figure 1)

Figure 1. Heterogeneity Assessment by Comparing Trial- and Patient-Level Characteristics Between T-02 and E-002 Trials

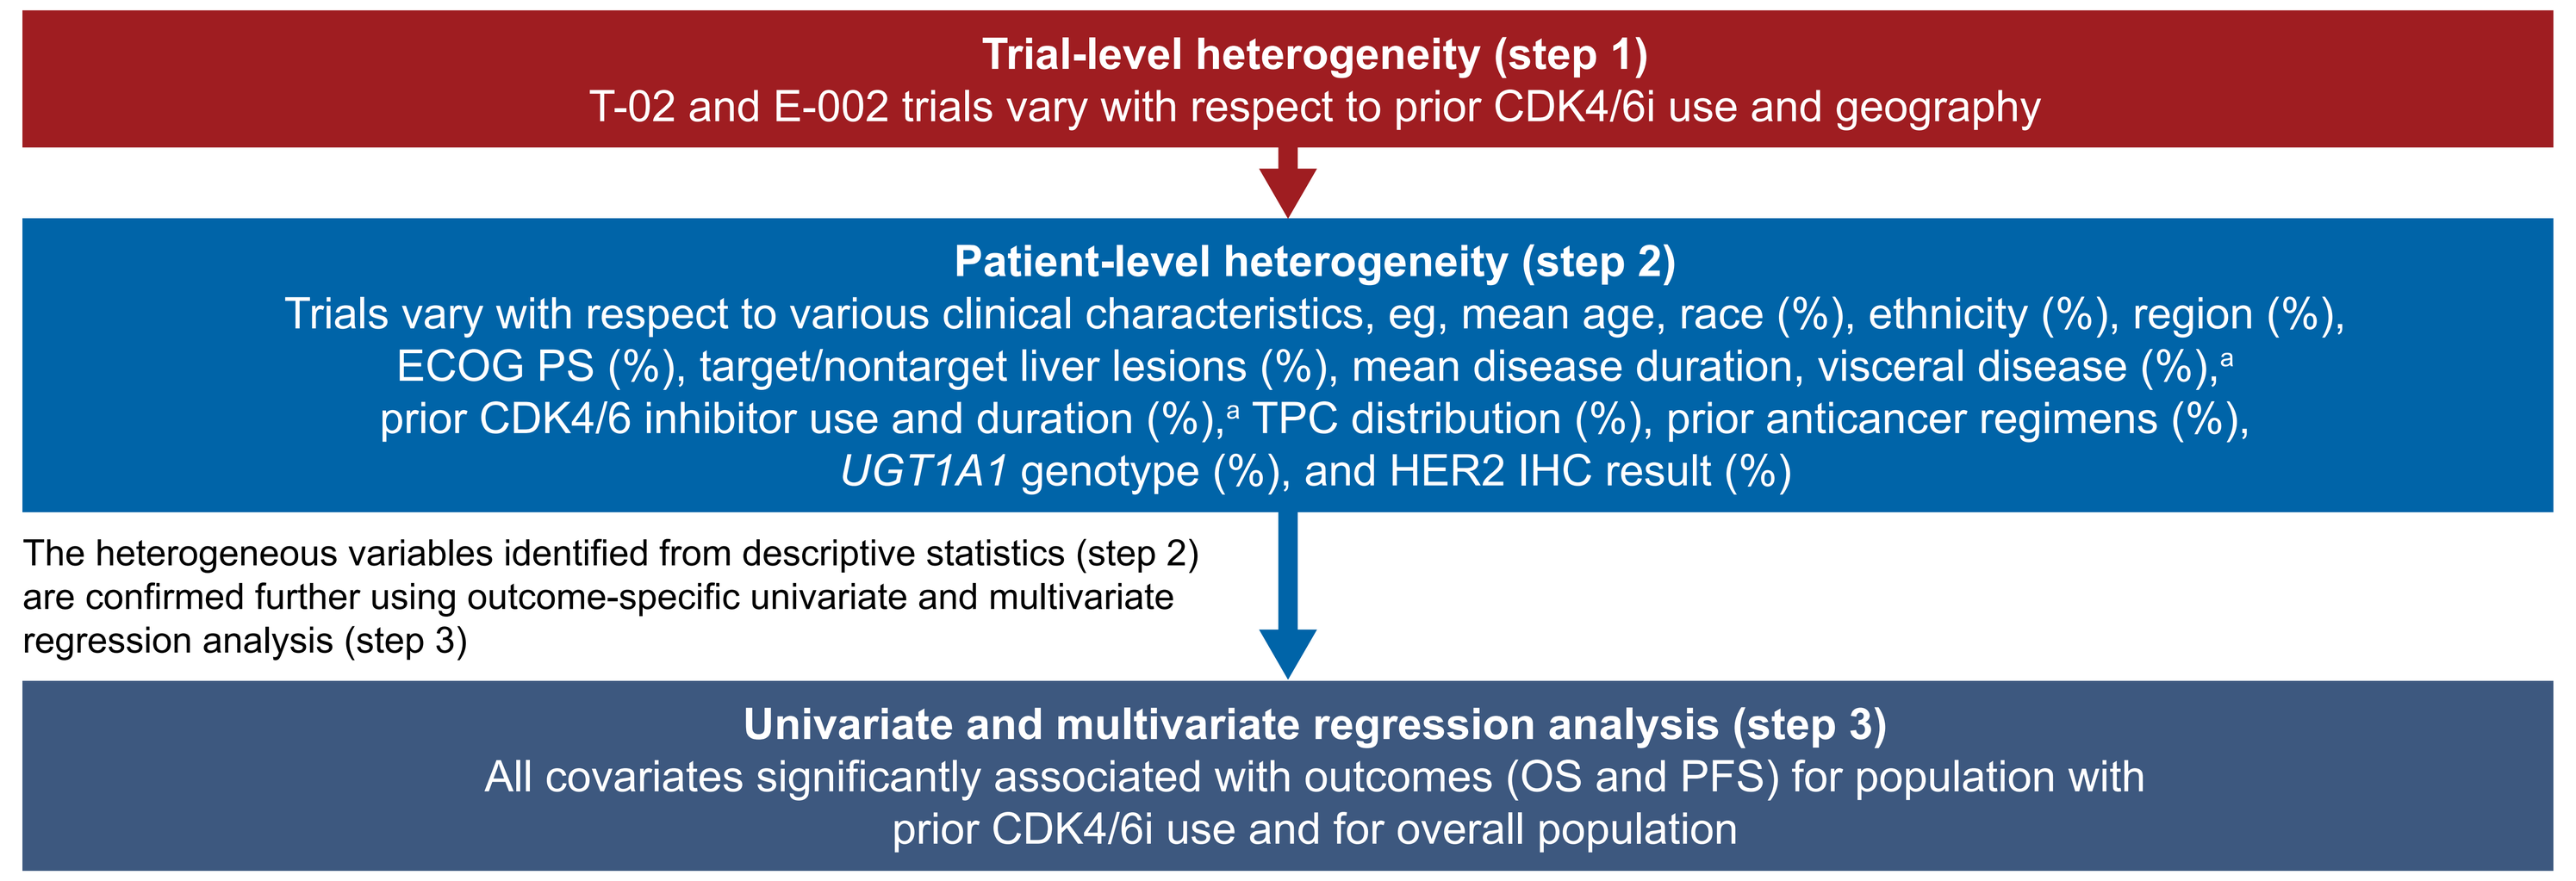

Trial-level heterogeneity (step 1)
T-02 and E-002 trials vary with respect to prior CDK4/6i use and geography
Patient-level heterogeneity (step 2)
Trials vary with respect to various clinical characteristics, eg, mean age, race (%), ethnicity (%), region (%), ECOG PS (%), target/nontarget liver lesions (%), mean disease duration, visceral disease (%),a prior CDK4/6 inhibitor use and duration (%),a TPC distribution (%), prior anticancer regimens (%), UGT1A1 genotype (%), and HER2 IHC result (%)
The heterogeneous variables identified from descriptive statistics (step 2) are confirmed further using outcome-specific univariate and multivariate regression analysis (step 3)
Univariate and multivariate regression analysis (step 3)
All covariates significantly associated with outcomes (OS and PFS) for population with prior CDK4/6i use and for overall population

[a]Selected based on clinical recommendations.
CDK4/6i, cyclin-dependent kinase 4/6 inhibitor; E-002, EVER-132-002; ECOG PS, Eastern Cooperative Oncology Group performance status; HER2, human epidermal growth factor receptor-2; IHC, immunohistochemistry; OS, overall survival; PFS, progression-free survival; T-02, TROPiCS-02; TPC, treatment of physician's choice; *UGT1A1*, uridine diphosphate glucuronosyltransferase.

Results

Baseline Characteristics

- In general, T-02 and E-002 had similar distribution of baseline population characteristics (low heterogeneity, I^2 = 0.0%-9.6%) except for prior CDK4/6i treatment and geography

Outcomes

- Median OS was 16.2 months (95% CI, 14.9-18.1) and 12.7 months (95% CI, 11.5-13.9) for SG and TPC, respectively, in the overall population. Results were similar for patients who received prior CDK4/6i treatment (Figure 2)
- In the fixed-effect model and one-stage approach, SG showed statistically significant improvement vs TPC in OS (Table 1; Figure 3) and PFS (Table 2; Figure 4), both for patients with prior CDK4/6i use and for the overall population

Figure 2. Overall Survival With SG and TPC in the Overall Population (A) and Patients With Prior CDK4/6i (B) From the Combined T-02 and E-002 Trials

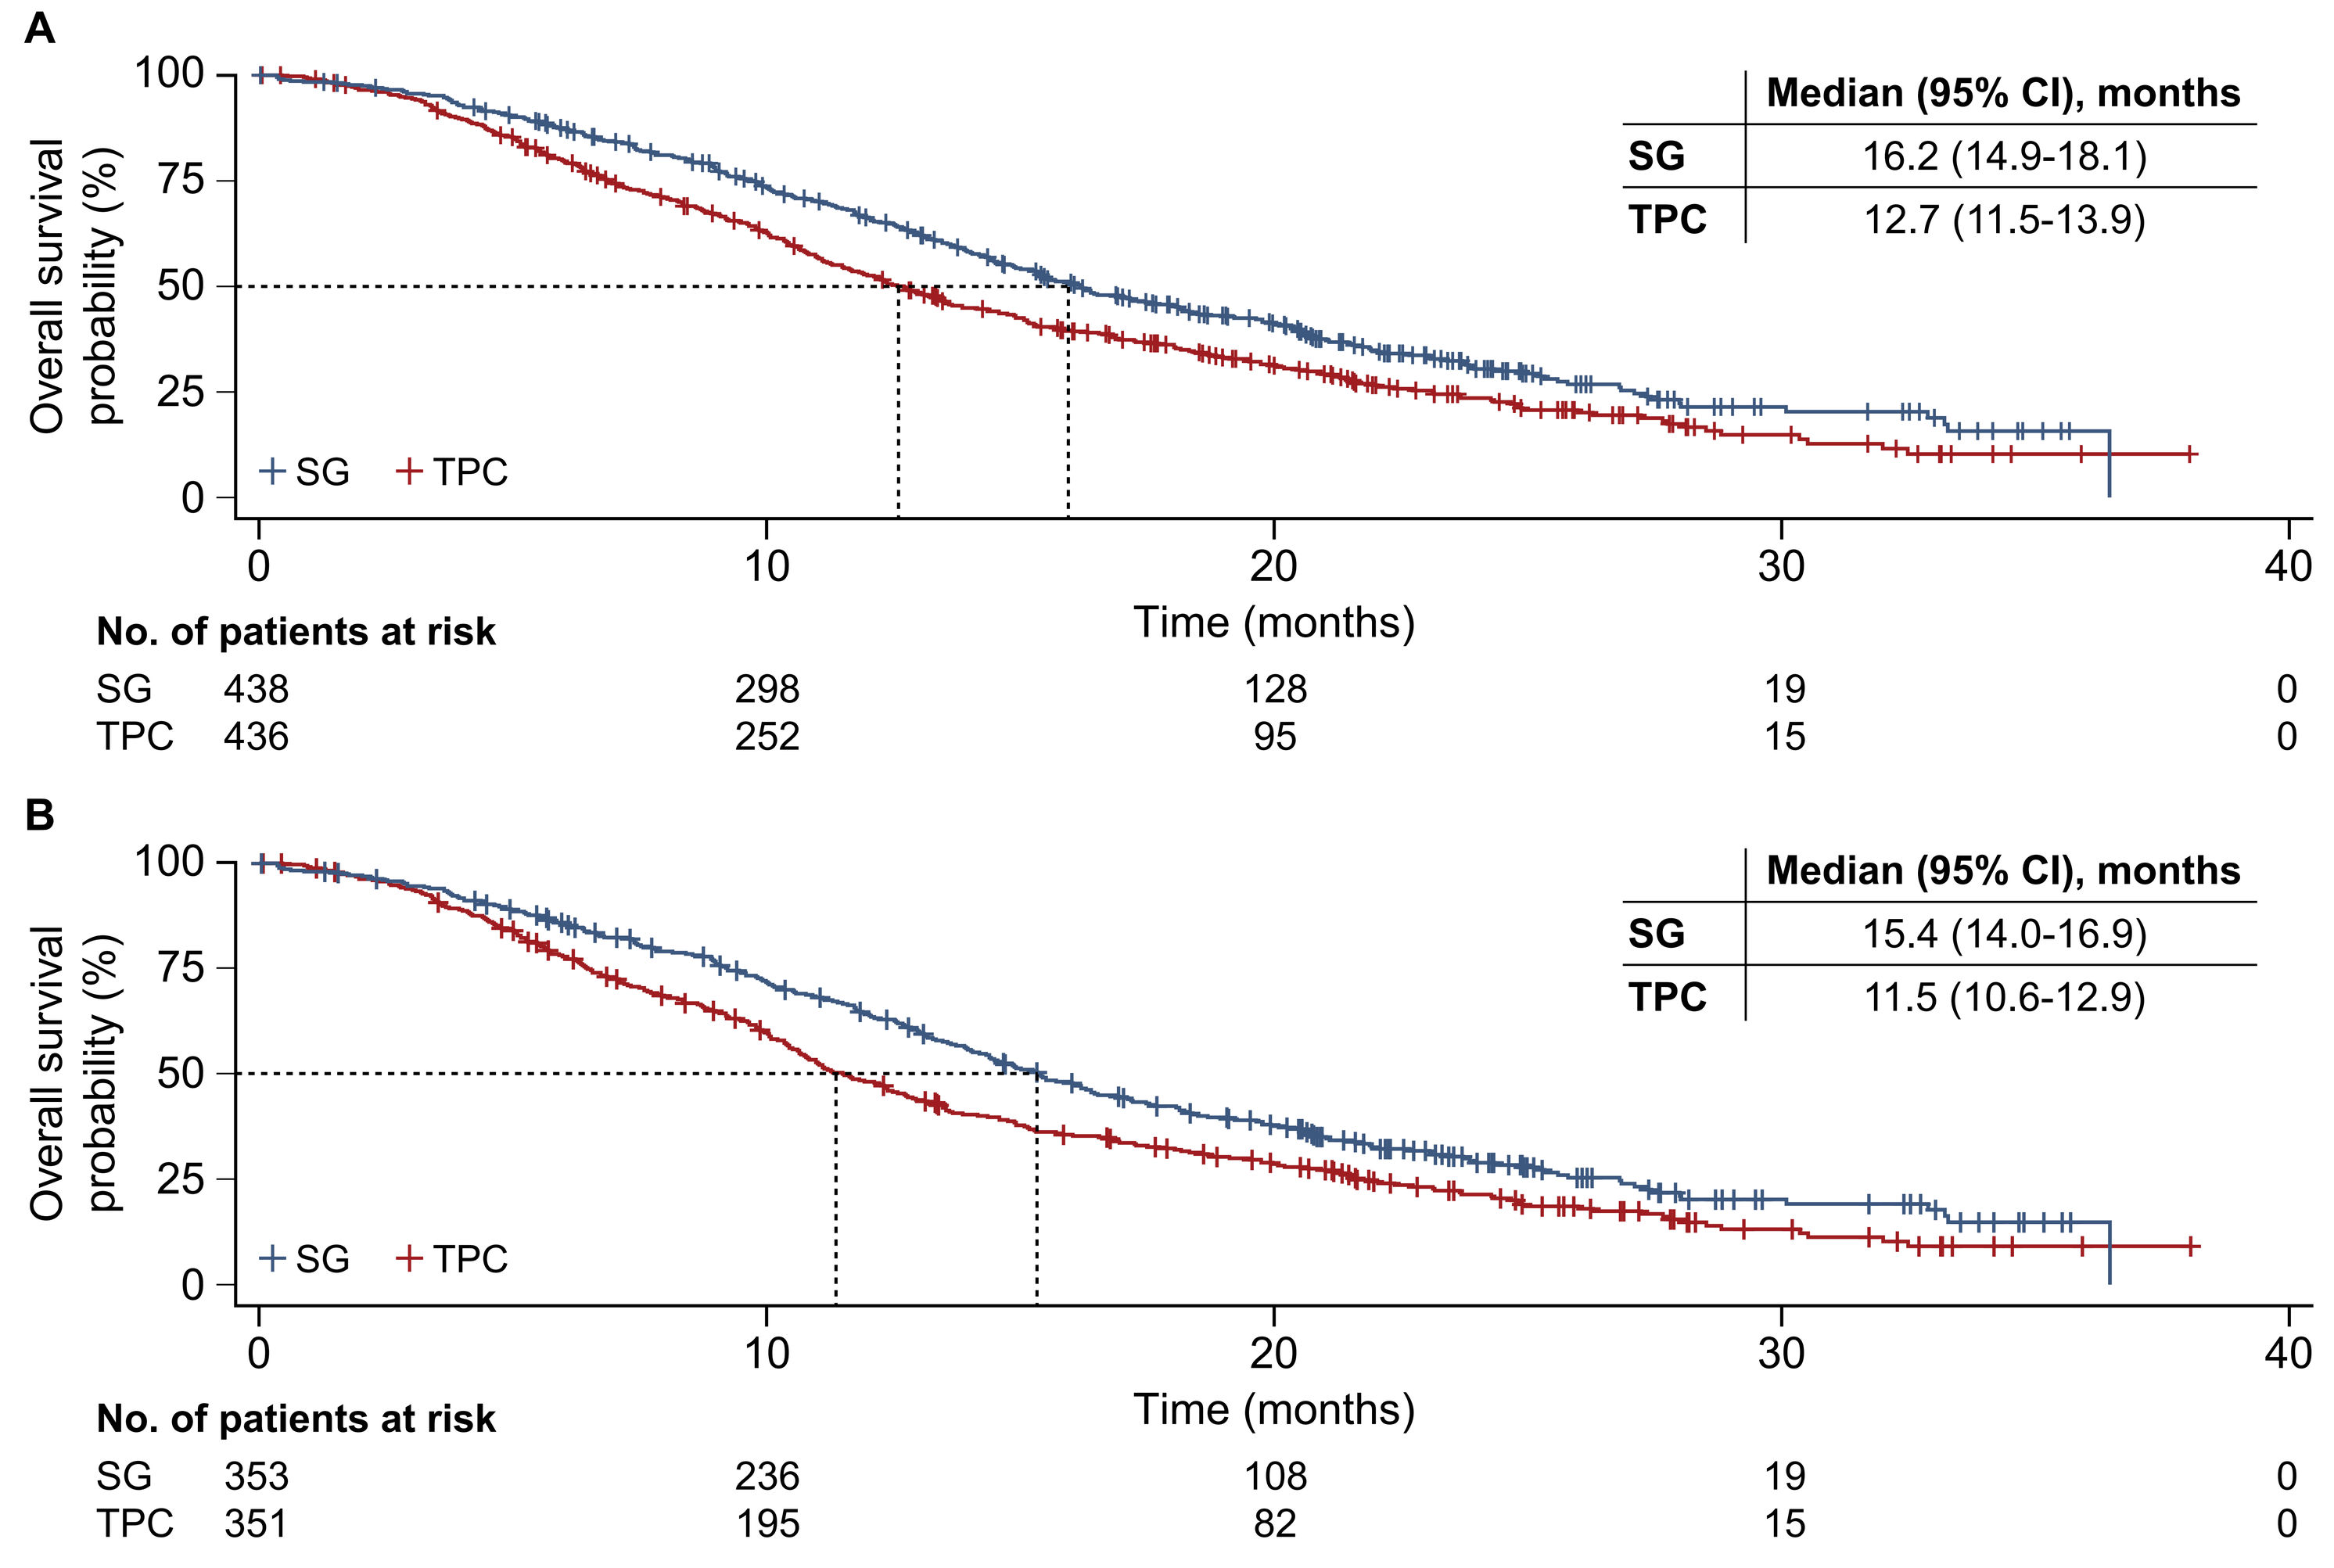

A
Overall survival probability (%)
100
75
50
25
0
+SG +TPC
0
10
20
30
40
Time (months)
Median (95% CI), months
SG 16.2 (14.9-18.1)
TPC 12.7 (11.5-13.9)
No. of patients at risk
SG 438 298 128 19 0
TPC 436 252 95 15 0
B
Median (95% CI), months
SG 15.4 (14.0-16.9)
TPC 11.5 (10.6-12.9)
No. of patients at risk
SG 353 236 108 19 0
TPC 351 195 82 15 0

CDK4/6i, cyclin-dependent kinase 4/6 inhibitor; E-002, EVER-132-002; SG, sacituzumab govitecan; T-02, TROPiCS-02; TPC, treatment of physician's choice.

Table 1. Comparison of Overall Survival With SG vs TPC in the Overall Population and Patients With Prior CDK4/6i From the Combined T-02 and E-002 Trials (Fixed-Effect Model and One-Stage Approach)

Population	HR (95% CI) for SG vs TPC	I^2 Statistic[a]	Q (Chi² Statistic)[a]
Overall population (N = 874)			
Fixed-effect model[b]	0.70 (0.58-0.86), *P* < .001	9.6% (no/low)	1.11, *P* = .29
One-stage approach	0.66 (0.55-0.80), *P* < .001	-	-
Prior CDK4/6i use (n = 704)			
Fixed-effect model[b]	0.68 (0.55-0.84), *P* < .001	73.8% (moderate)	3.82, *P* = .05
One-stage approach	0.65 (0.53-0.80), *P* < .001	-	-

Results based on statistical and clinical covariates.
[a]I^2 and Q (Chi²) statistics are measures for evaluating the level of heterogeneity between studies and hence relevant in the fixed-effect model analysis; the higher the I^2 value, the higher the heterogeneity of the studies; a significant Chi² value indicates the presence of heterogeneity between the studies.
[b]The fixed-effect model assumes that the studies share a single common effect, and therefore, all the variance in the observed effect is attributable to sampling error.
CDK4/6i, cyclin-dependent kinase 4/6 inhibitor; E-002, EVER-132-002; HR, hazard ratio; SG, sacituzumab govitecan; T-02, TROPiCS-02; TPC, treatment of physician's choice.

Figure 3. Comparison of Overall Survival With SG vs TPC in the Overall Population (A) and Patients With Prior CDK4/6i (B) From T-02 and E-002 Trials (Fixed-Effect Model)

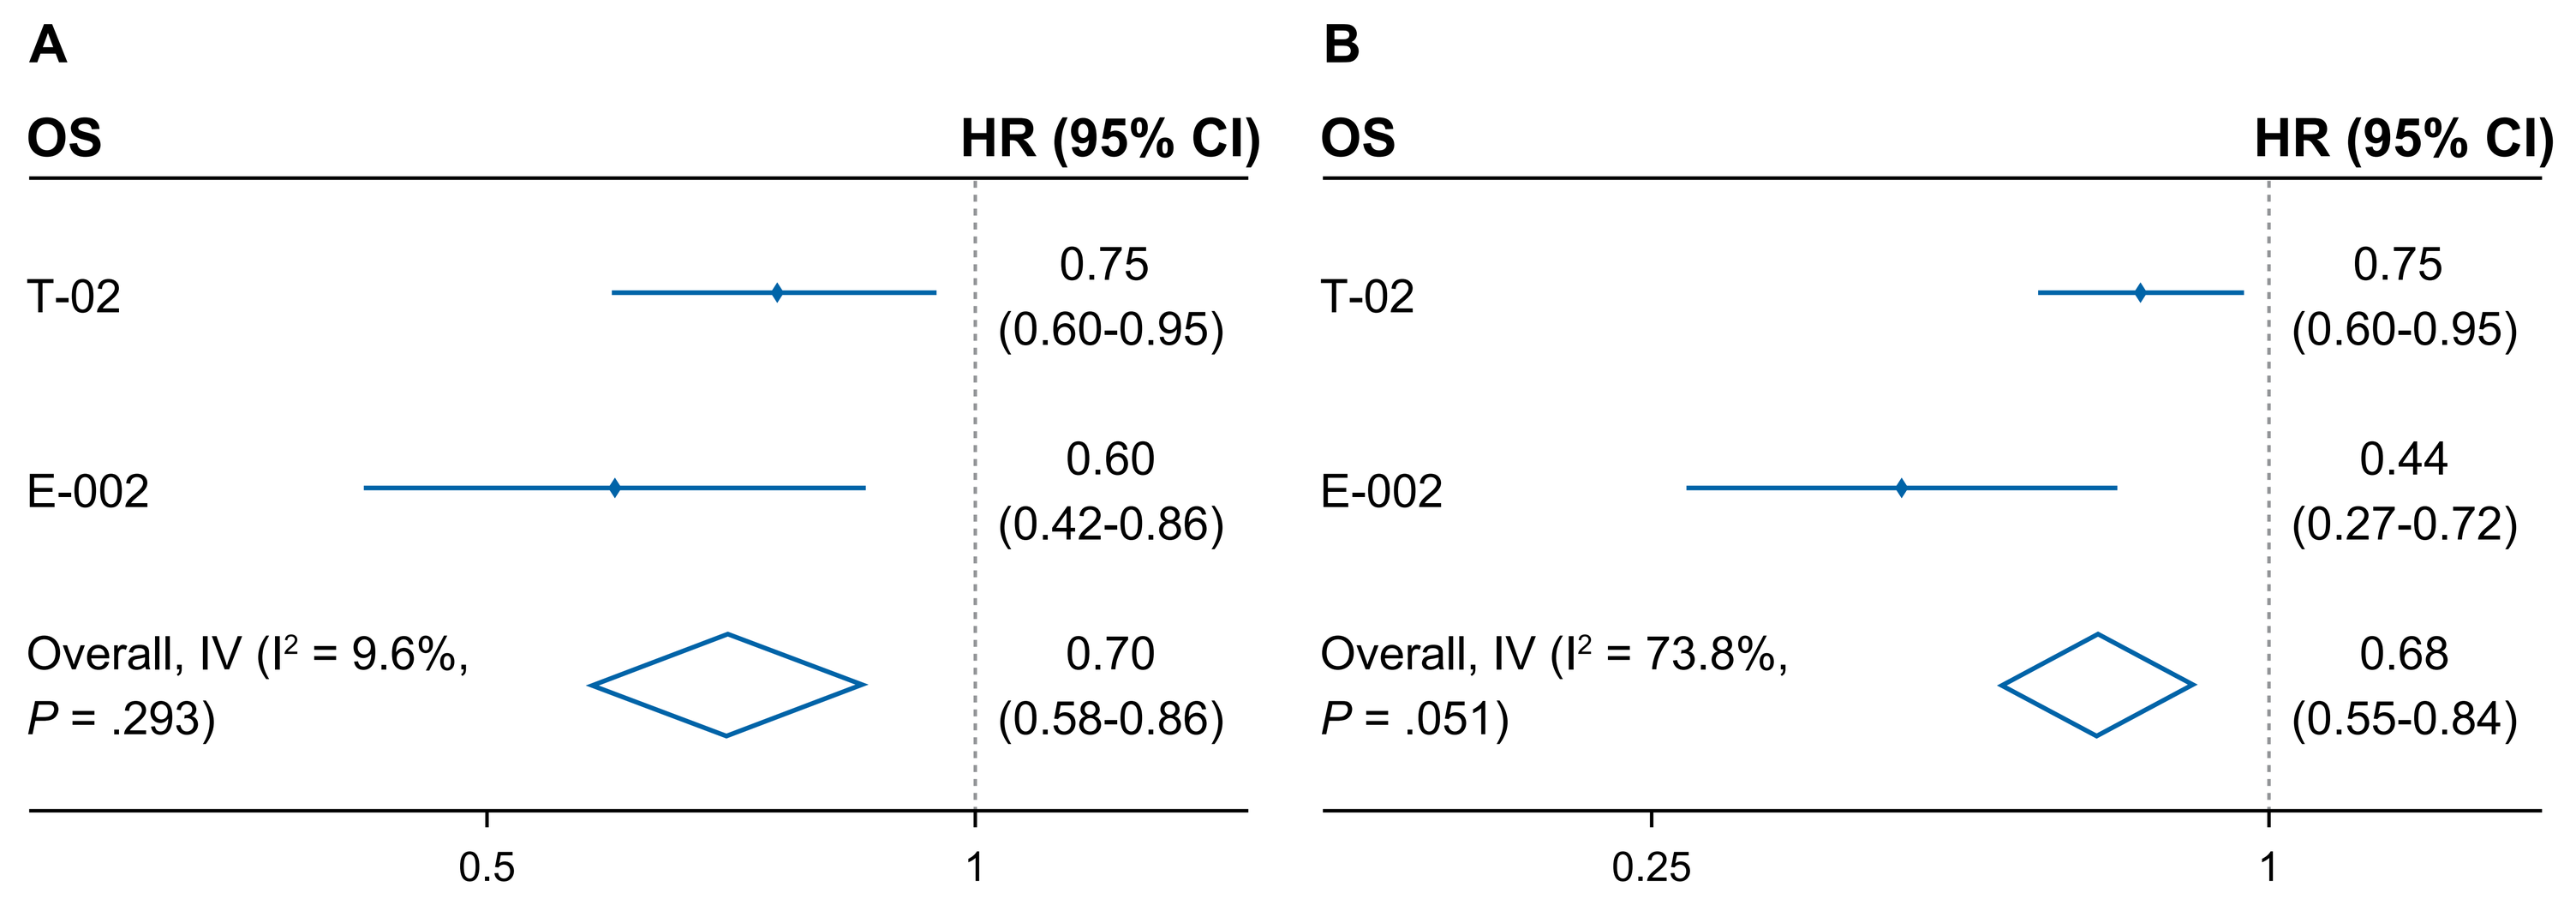

A
OS
HR (95% CI)
T-02
0.75 (0.60-0.95)
E-002
0.60 (0.42-0.86)
Overall, IV (I² = 9.6%, P = .293)
0.70 (0.58-0.86)
0.5
1
B
OS
HR (95% CI)
T-02
0.75 (0.60-0.95)
E-002
0.44 (0.27-0.72)
Overall, IV (I² = 73.8%, P = .051)
0.68 (0.55-0.84)
0.25
1

Results based on statistical and clinical covariates.
CDK4/6i, cyclin-dependent kinase 4/6 inhibitor; E-002, EVER-132-002; HR, hazard ratio; IV, inverse variance; OS, overall survival; SG, sacituzumab govitecan; T-02, TROPiCS-02; TPC, treatment of physician's choice.

Table 2. Comparison of Progression-Free Survival With SG vs TPC in the Overall Population and Patients With Prior CDK4/6i From the Combined T-02 and E-002 Trials (Fixed-Effect Model and One-Stage Approach)

Population	HR (95% CI) for SG vs TPC	I^2 Statistic[a]	Q (Chi² Statistic)[a]
Overall population (N = 874)			
Fixed-effect model[b]	0.67 (0.55-0.83), *P* < .001	0.0% (No)	0.38, *P* = .54
One-stage approach	0.62 (0.50-0.77), *P* < .001	-	-
Prior CDK4/6i use (n = 704)			
Fixed-effect model[b]	0.66 (0.52-0.84), *P* = .001	0.0% (No)	0.89, *P* = .35
One-stage approach	0.65 (0.52-0.81), *P* < .001	-	-

Results based on statistical and clinical covariates.
[a]I^2 and Q (Chi²) statistics are measures for evaluating the level of heterogeneity between studies and hence relevant in the fixed-effect model analysis; the higher the I^2 value, the higher the heterogeneity of the studies; a significant Chi² value indicates the presence of heterogeneity between the studies.
[b]Fixed-effect model assumes the studies share a single common effect, and therefore, all the variance in the observed effect is attributable to sampling error.
CDK4/6i, cyclin-dependent kinase 4/6 inhibitor; E-002, EVER-132-002; HR, hazard ratio; SG, sacituzumab govitecan; T-02, TROPiCS-02; TPC, treatment of physician's choice.

Figure 4. Comparison of Progression-Free Survival With SG vs TPC in the Overall Population (A) and Patients With Prior CDK4/6i (B) From T-02 and E-002 Trials (Fixed-Effect Model)

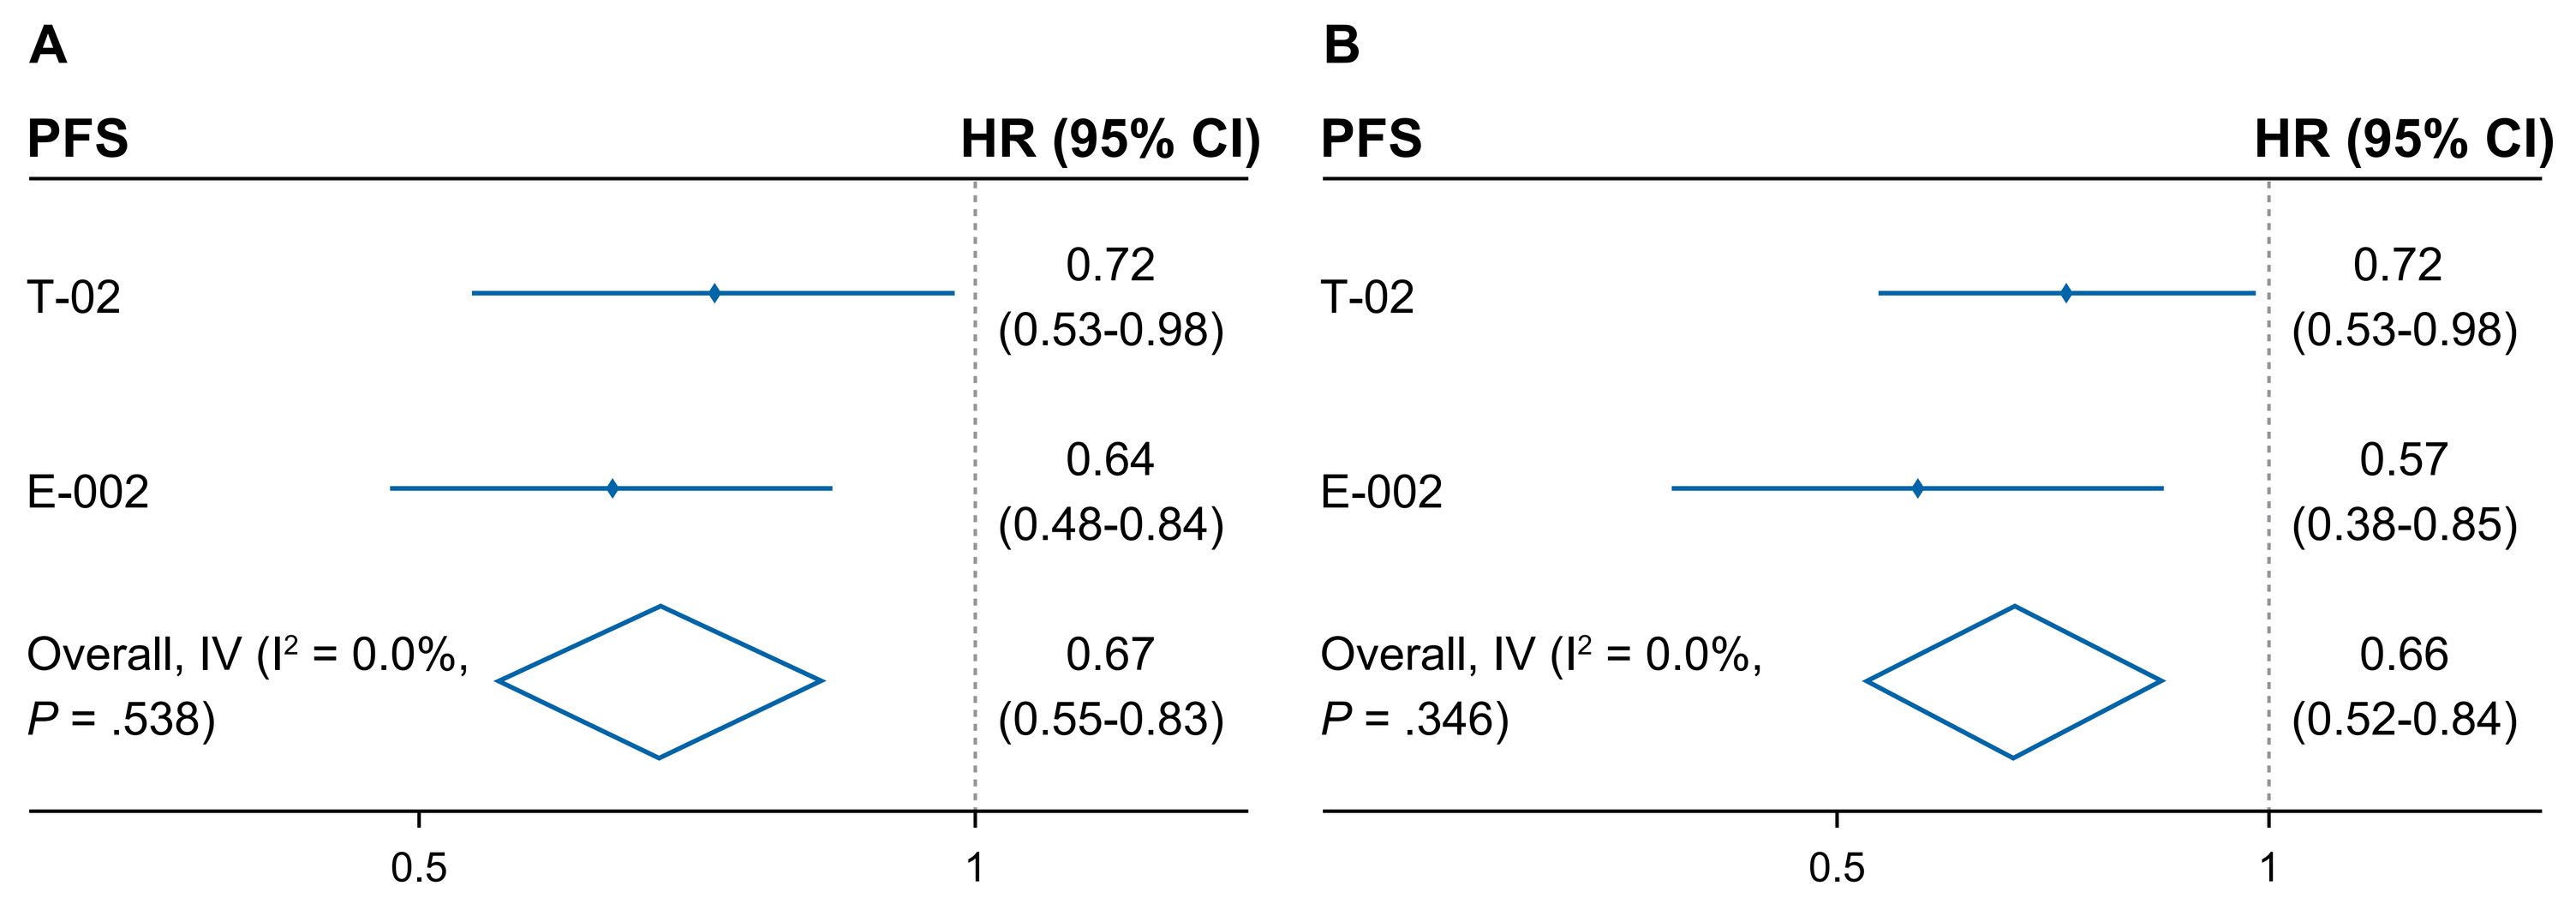

A
PFS
HR (95% CI)
T-02
0.72 (0.53-0.98)
E-002
0.64 (0.48-0.84)
Overall, IV (I² = 0.0%, P = .538)
0.67 (0.55-0.83)
0.5
1
B
PFS
HR (95% CI)
T-02
0.72 (0.53-0.98)
E-002
0.57 (0.38-0.85)
Overall, IV (I² = 0.0%, P = .346)
0.66 (0.52-0.84)
0.5
1

Results based on statistical and clinical covariates.
CDK4/6i, cyclin-dependent kinase 4/6 inhibitor; E-002, EVER-132-002; HR, hazard ratio; IV, inverse variance; PFS, progression-free survival; SG, sacituzumab govitecan; T-02, TROPiCS-02; TPC, treatment of physician's choice.

References: 1. Starodub AN, et al. *Clin Cancer Res*. 2015;21;3870-78. 2. Rugo HS, et al. *J Clin Oncol*. 2022;40:3365-76. 3. Rugo H, et al. *Future Oncol*. 2020;12:705-15. 4. TRODELVY® (sacituzumab govitecan-hziy) [prescribing information]. Foster City, CA: Gilead Sciences, Inc., February 2023. 5. TRODELVY® (sacituzumab govitecan-hziy) [summary of product characteristics]. County Cork, Ireland: Gilead Sciences Ireland UC; August 2023. 6. Xu B, et al. *Ann Oncol*. 2023;34(suppl 4):S1485.

Acknowledgments: We extend our thanks to the patients, their families, and all participating investigators. This study was funded by Gilead Sciences, Inc. Medical writing and editorial assistance were provided by Peggy Robinet, PharmD, PhD, of Parexel, and funded by Gilead Sciences, Inc.

Presenting Author Disclosures: **OG** declares being on the advisory/speaker board of Roche, Lilly, Novartis, Pierre Fabre, MSD, AstraZeneca, Pfizer, Exact Sciences, Gilead Sciences, Inc., Seagen, and Agendia, and holding leadership role/proprietary information at West German Study Group.

Correspondence: Dr Hope Rugo, hope.rugo@ucsf.edu

ESMO Breast Cancer; May 15-17, 2024; Berlin, Germany